2511

Identifying Predictive Plasma Proteomic Biomarkers for Clinical Response to AXA1125, an Endogenous Metabolic Modulator (EMM) Composition Being Developed for the Treatment of NAFLD/NASH

Heping Wang, Matthew Russell, Joel Pradines, Cerasela Albei, and Karim Azer

Axcella Therapeutics, Cambridge, MA, United States

Introduction

- Proteomics and metabolomics data represent opportunities to identify stable and dynamic biomarkers that are measurable in biological fluids
- In nonalcoholic steatohepatitis (NASH), these biomarkers are being explored for classifying disease, monitoring disease progression, and assessing treatment response, using signatures representing different components of disease biologies (steatosis, inflammation, ballooning, fibrosis)[1,2]
- AXA1125, a novel endogenous metabolic modulator composition of 5 specific amino acids and N-acetylcysteine, was developed to simultaneously target pathways related to liver metabolism, inflammation, and fibrosis (Figure 1)[3]
- The AXA1125-003 study (NCT04073368), a multicenter, 16-week, single-blind study in subjects with nonalcoholic fatty liver disease (NAFLD) (Figure 1A), showed that AXA1125 had consistently greater effect than placebo in reducing magnetic resonance imaging-proton density fat fraction (MRI-PDFF), a measure of liver fat content (Figure 1B)[3]
- At Week 16, a greater proportion of subjects who received AXA1125 (35%–40%) versus placebo (8%–25%) achieved a ≥30% reduction in MRI-PDFF (Figure 1C),[3] a threshold associated with histopathologic improvement in NASH[4-6]
- AXA1125 is currently being studied in a phase 2b study (AXA1125-101) in subjects with NASH and fibrosis (NCT04880187)
- As a part of our systems biology platform,[7] we explored stable plasma protein biomarkers that might predict clinically relevant treatment effects with AXA1125 in NAFLD/NASH

FIGURE 1

AXA1125-003 Study Design (A), Results of MRI-PDFF Assessments Showing Mean Change From Baseline in MRI-PDFF (B), and Proportion of Subjects With ≥30% Reduction in MRI-PDFF (C)

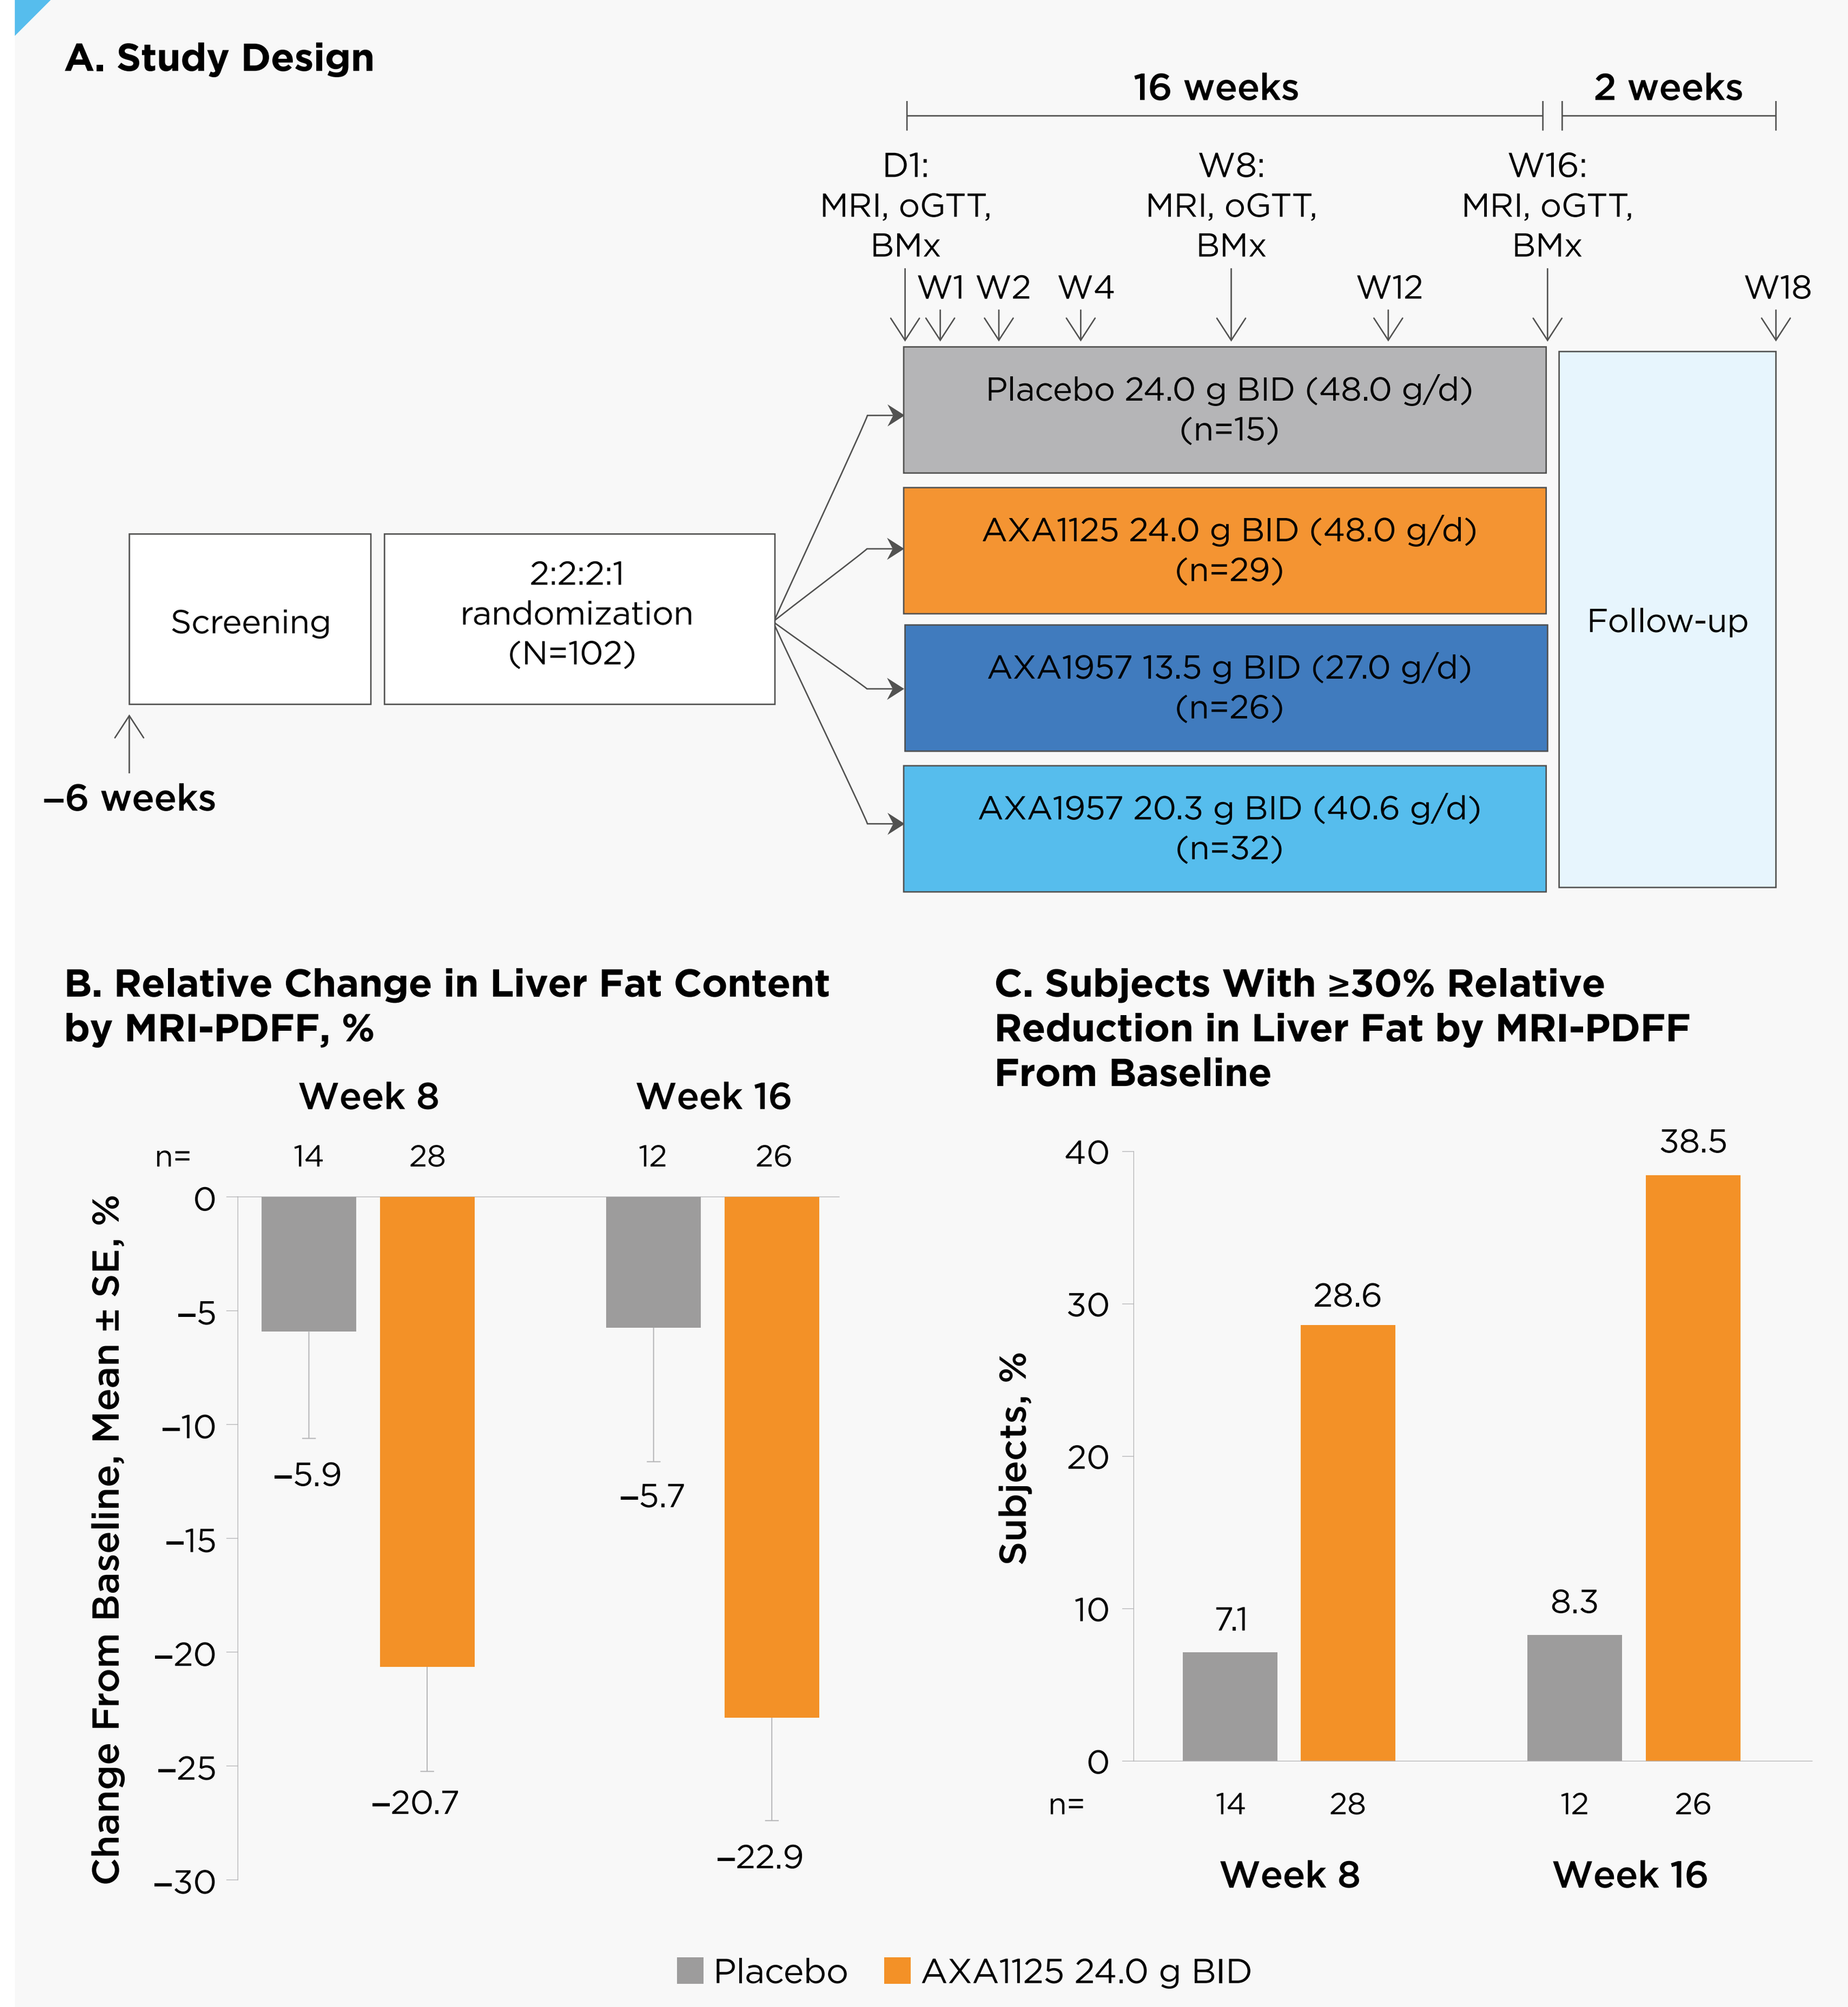
A. Study Design
16 weeks
2 weeks
D1: MRI, oGTT, BMx
W8: MRI, oGTT, BMx
W16: MRI, oGTT, BMx
W1 W2 W4
W12
W18
Screening
2:2:2:1 randomization (N=102)
Placebo 24.0 g BID (48.0 g/d) (n=15)
AXA1125 24.0 g BID (48.0 g/d) (n=29)
AXA1957 13.5 g BID (27.0 g/d) (n=26)
AXA1957 20.3 g BID (40.6 g/d) (n=32)
Follow-up
−6 weeks
B. Relative Change in Liver Fat Content by MRI-PDFF, %
Week 8
Week 16
n= 14 28 12 26
Change From Baseline, Mean ± SE, %
0 −5 −10 −15 −20 −25 −30
−5.9
−20.7
−5.7
−22.9
C. Subjects With ≥30% Relative Reduction in Liver Fat by MRI-PDFF From Baseline
Subjects, %
40 30 20 10 0
7.1
28.6
8.3
38.5
n= 14 28 12 26
Week 8
Week 16
Placebo
AXA1125 24.0 g BID

Reproduced from Harrison SA, et al.[3]

BID, twice daily; BMx, biomarkers; D, day; MRI, magnetic resonance imaging; MRI-PDFF, magnetic resonance imaging-proton density fat fraction; oGTT, oral glucose tolerance test; SE, standard error; W, week.

Aim

- To identify biomarkers linked to the MRI-PDFF reduction observed with AXA1125 treatment in NAFLD/NASH
- To demonstrate the feasibility of predicting which subjects will be the most responsive to AXA1125 treatment
- To identify ways of selecting and stratifying subjects in future trials of AXA1125

Methods

- SomaScan® Discovery proteomics (7378 proteins) was performed on the plasma samples collected at baseline (Day 1), Week 8, and Week 16 from subjects treated with AXA1125 and with placebo in the AXA1125-003 study
- After removal of quality control–flagged samples, proteomics data were available for 24, 25, and 24 subjects treated with AXA1125 and 12, 12, and 11 subjects treated with placebo on Day 1, Week 8, and Week 16, respectively
- Proteomic data, demographics, and baseline clinical characteristics (age, height, waist circumference, body mass index [BMI], and type 2 diabetes status) were considered as explanatory variables (Table 1)
 - Potential variables were selected for their close association with change in MRI-PDFF, based on Spearman's rank correlation
 - Correlation analysis was performed to identify potential biomarkers ($P<0.05$) specific to the AXA1125 treatment group
- MRI-PDFF change from baseline to Weeks 8 and 16 represented the response variable
 - Responses were categorized as either ≥30% or <30% reduction in MRI-PDFF

Table 1: Demographics and Baseline Clinical Characteristics of the AXA1125 and Placebo Treatment Arms of the AXA1125-003 Study[3]

	Placebo (n=15)	AXA1125 24 g BID (n=29)
Age (SE), years	53.2 (2.48)	49.2 (2.38)
Female, n (%)	10 (66.7)	17 (58.6)
Weight, kg	118.25 (8.20)	102.86 (4.42)
BMI (SE), kg/m²	42.0 (2.42)	36.8 (1.36)
Type 2 diabetes, n (%)	6 (40.0)	12 (41.4)
HbA1c, %	6.9	7.8
Metabolism		
MRI-PDFF, %	21.19 (1.51)	22.35 (0.93)
HOMA-IR	8.59 (1.26)	13.51 (3.39)
Inflammation		
ALT, U/L	50.5 (8.73)	55.2 (4.89)
AST, U/L	41.3 (34.29)	37.3 (22.69)
cT1, mSec	1022.3 (41.35)	960.6 (16.84)
CK-18 M65, U/L	900.5 (369.71)	450.8 (99.39)
Fibrosis		
FibroScan score, kPa (SE)	13.51 (1.47)	11.73 (1.24)
ProC3, ng/mL	15.85 (1.81)	17.07 (1.56)
ELF score	9.42 (0.32)	9.24 (0.17)
FIB-4	1.42 (0.25)	1.05 (0.12)

ALT, alanine aminotransferase; AST, aspartate aminotransferase; BID, twice daily; BMI, body mass index; CK-18 M65, creatine keratin-18 measured using M65 enzyme-linked immunosorbent assay; cT1, corrected T1; ELF, enhanced liver fibrosis; FIB-4, fibrosis-4; HbA1c, glycated hemoglobin; HOMA-IR, homeostasis model assessment of insulin resistance; MRI-PDFF, magnetic resonance imaging-proton density fat fraction; ProC3, N-terminal type III collagen propeptide; SE, standard error.

- A multiple logistic regression model was used to predict which subjects are likely to achieve greater reduction in MRI-PDFF after AXA1125 treatment

 PDFF response ~ baseline protein expression + demographic feature
- Differences in biomarker distribution between the 2 MRI-PDFF response categories (≥30% vs <30% reduction) were assessed with Wilcoxon's sum rank test, with statistical significance set at $P\leq0.05$
- The plasma proteins most closely associated with ΔPDFF were identified
- Cross-validation methods were conducted to assess regression model performance

Results

- Subjects with ≥30% MRI-PDFF reduction at Week 8 had significantly higher angiotensin converting enzyme-2 and serine protease 57 expression and lower baseline BMI than subjects with <30% MRI-PDFF reduction ($P<0.05$; Figure 2)
 - BMI correlated strongly with baseline waist circumference and weight

FIGURE 2

Selected Biomarkers and Week 8 MRI-PDFF Response

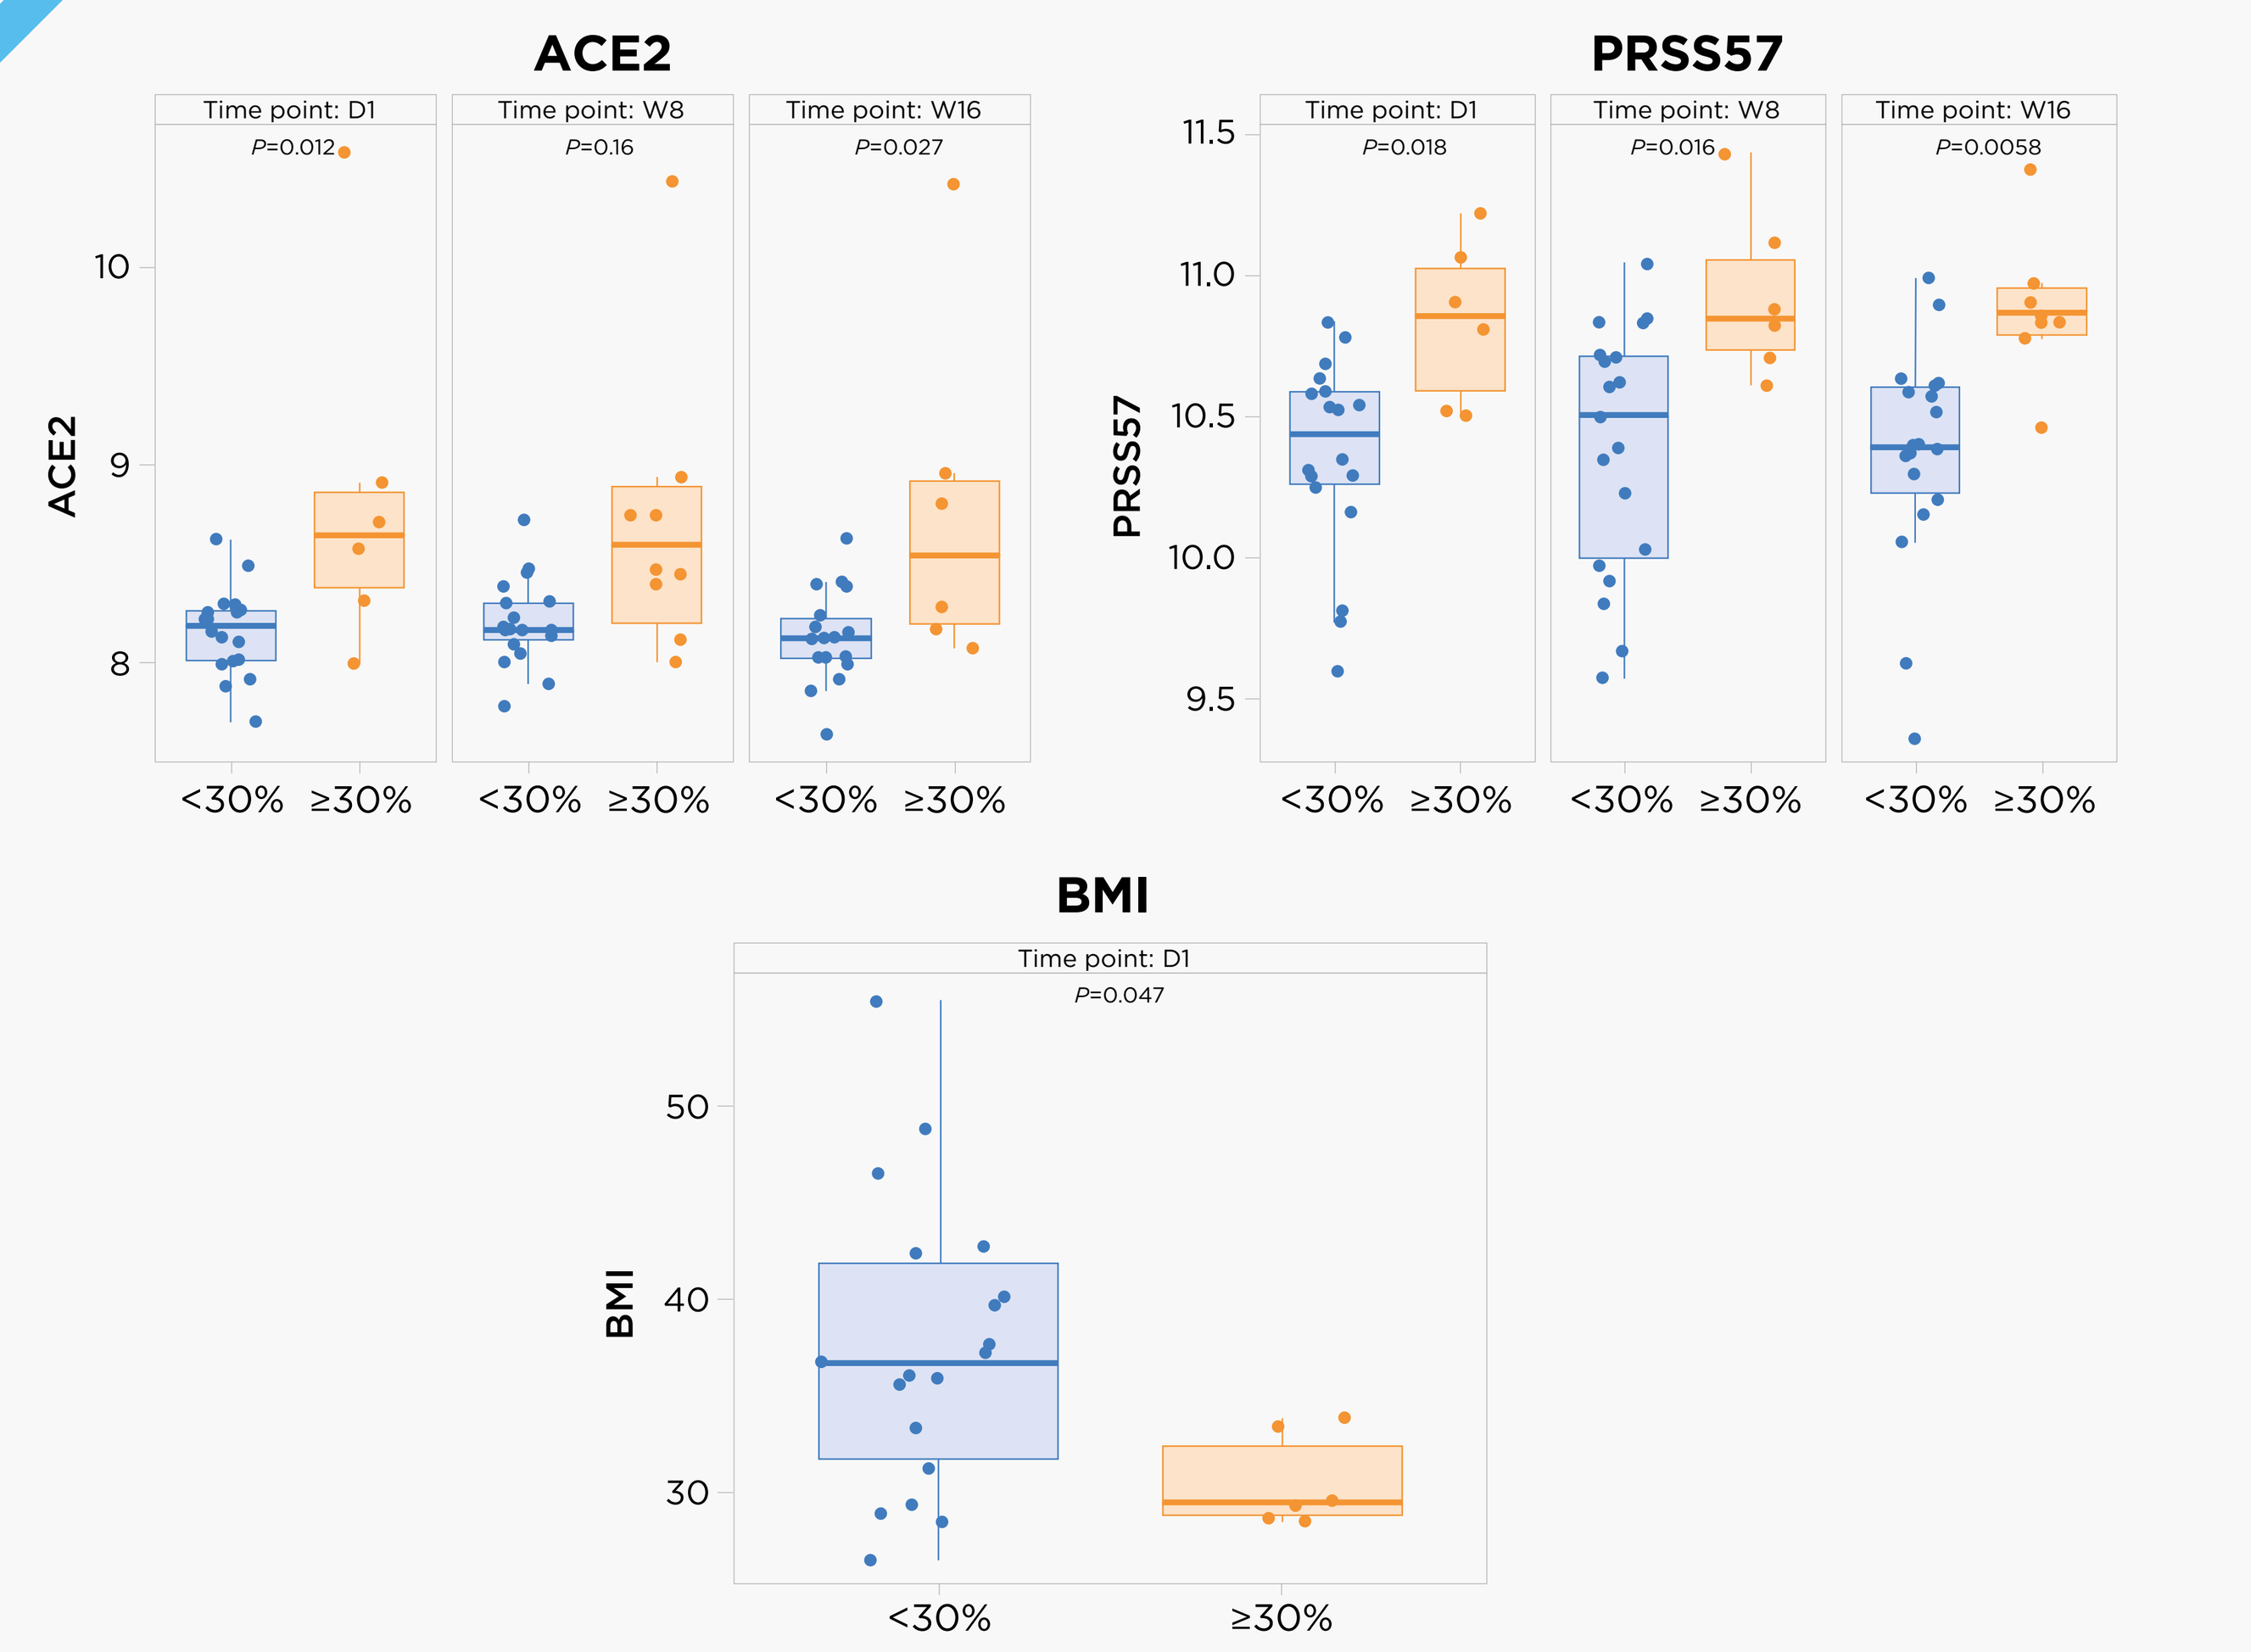
ACE2
Time point: D1 P=0.012
Time point: W8 P=0.16
Time point: W16 P=0.027
PRSS57
Time point: D1 P=0.018
Time point: W8 P=0.016
Time point: W16 P=0.0058
BMI
Time point: D1 P=0.047
<30% ≥30%

ACE2, angiotensin converting enzyme 2; BMI, body mass index; D, day; PRSS57, serine protease 57; W, week.

- Subjects with a ≥30% reduction in MRI-PDFF at Week 16 had a significantly lower expression of phosphatase and tensin homolog, higher expression of melanoregulin, and lower baseline BMI than subjects with a <30% reduction in MRI-PDFF ($P<0.05$; Figure 3)

FIGURE 3

Selected Biomarkers and Week 16 MRI-PDFF Response

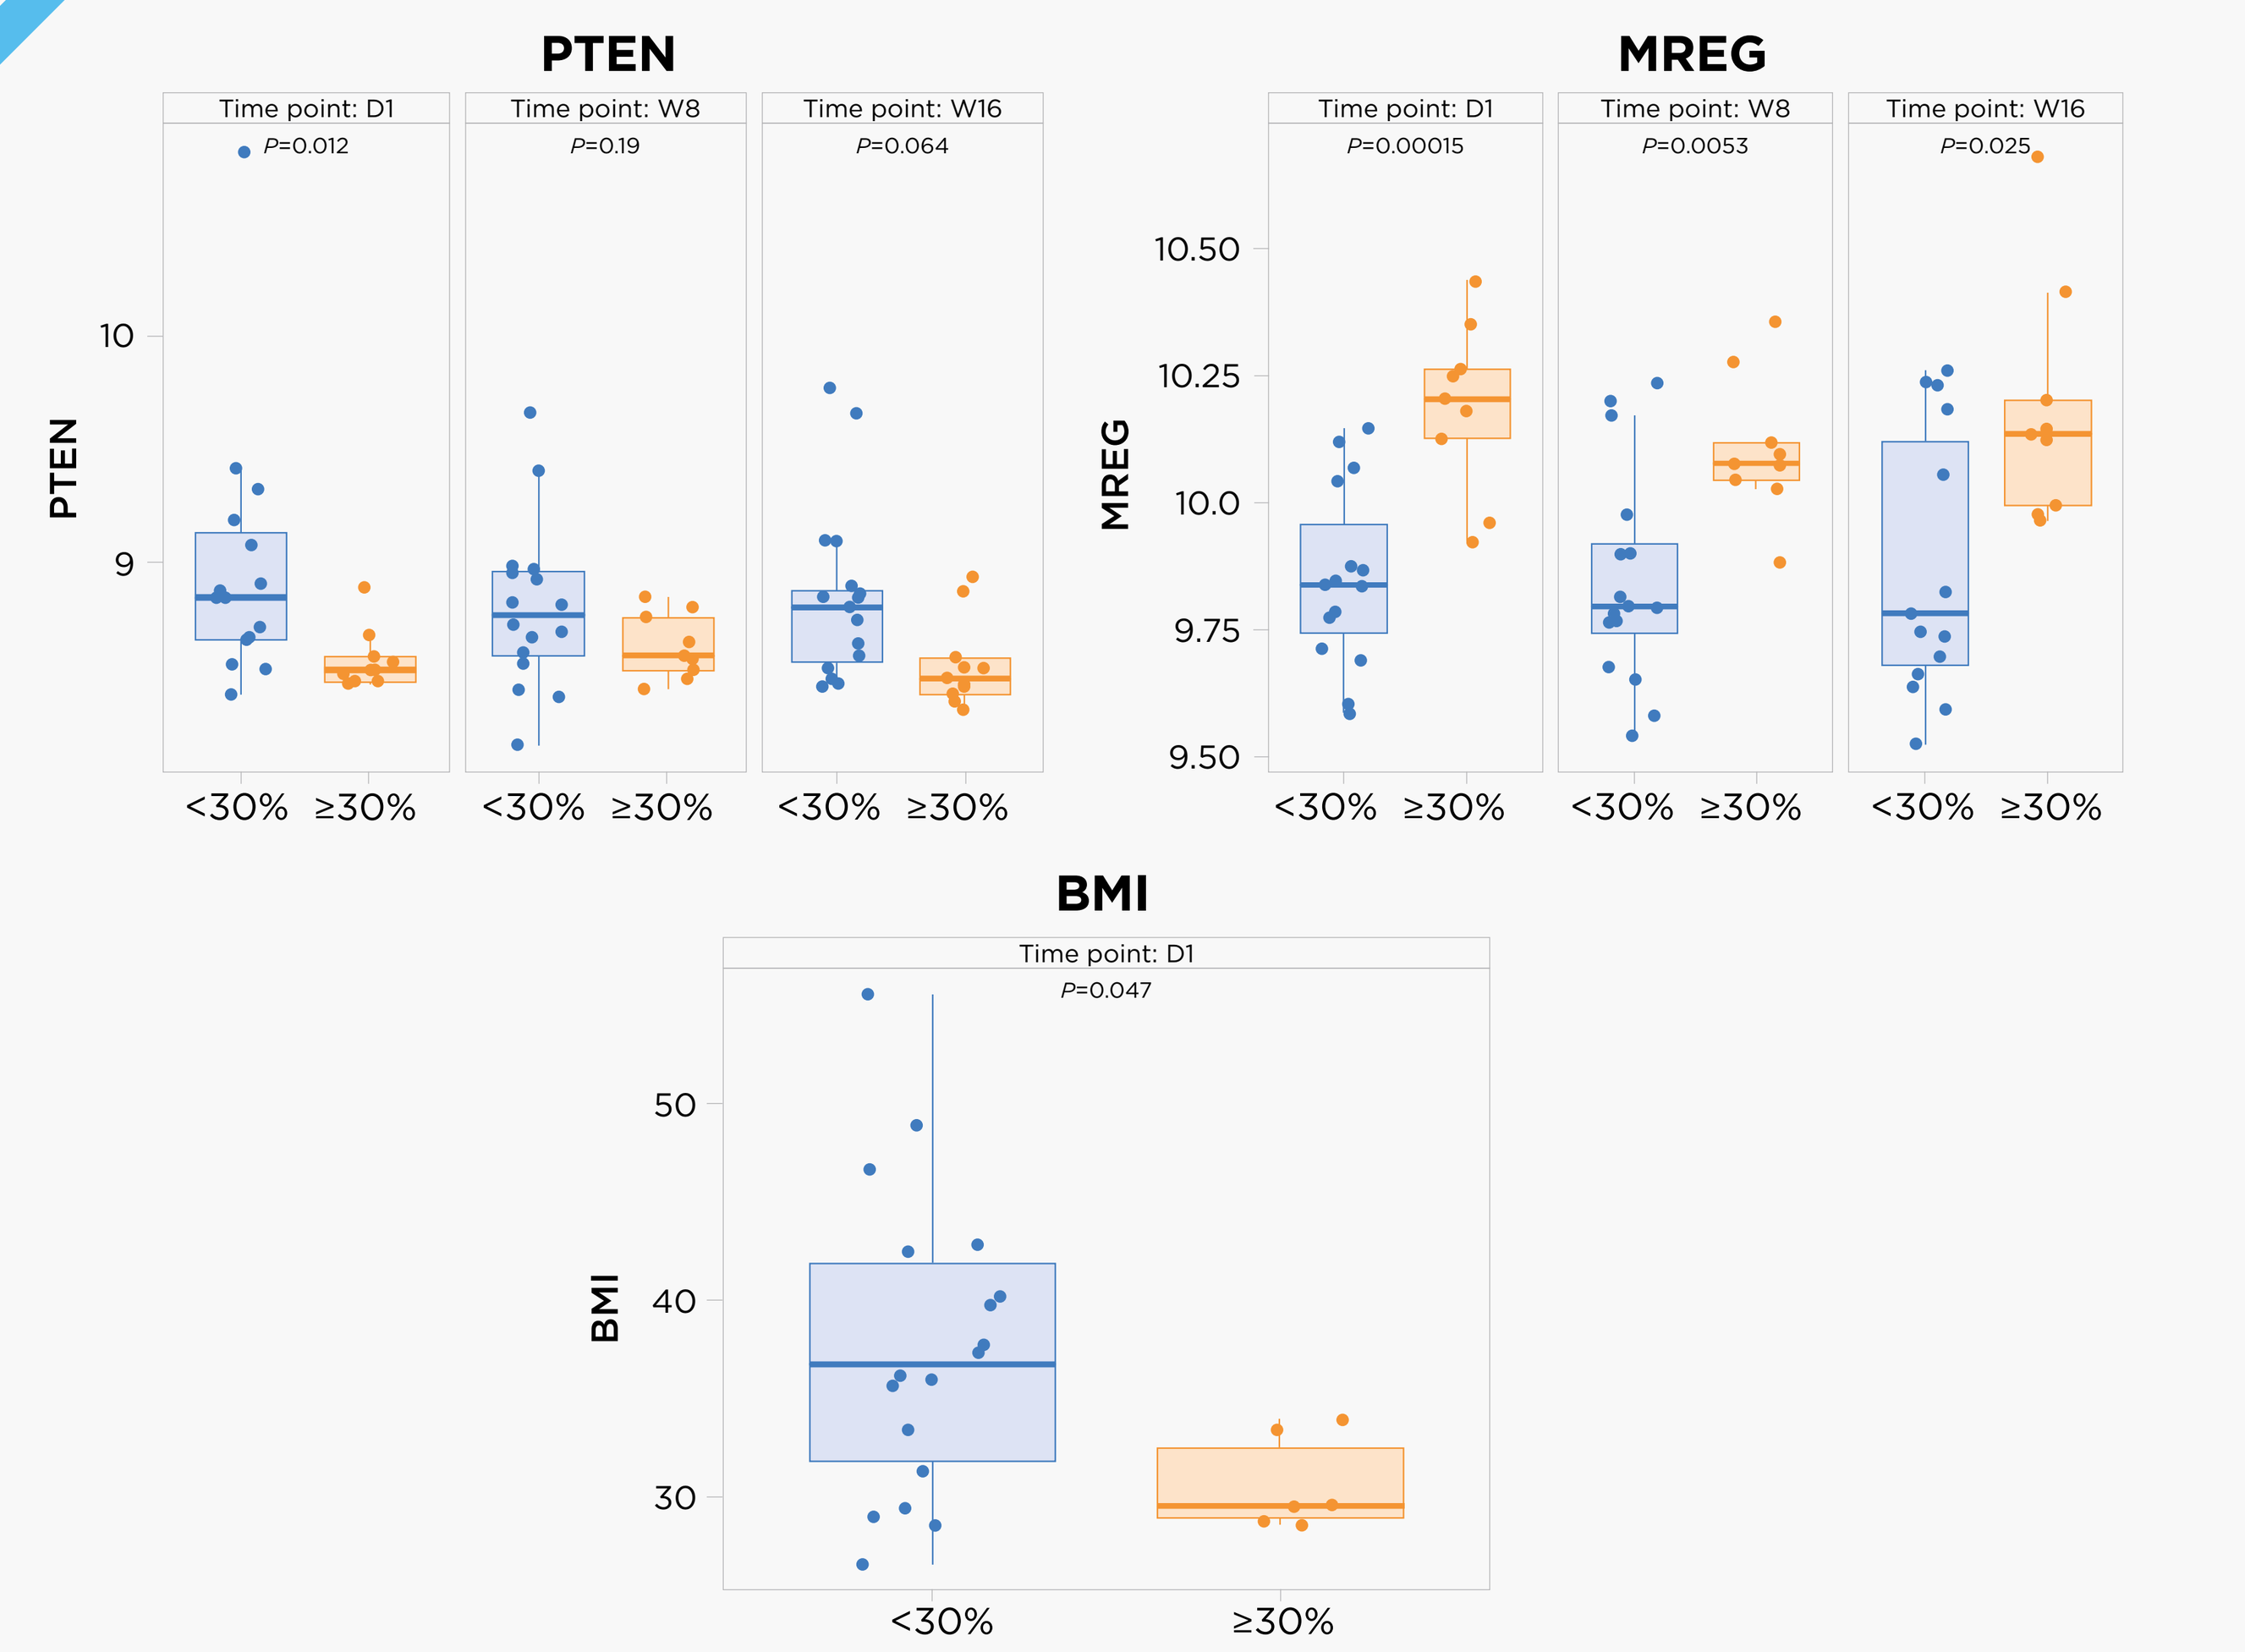
PTEN
Time point: D1 P=0.012
Time point: W8 P=0.19
Time point: W16 P=0.064
MREG
Time point: D1 P=0.00015
Time point: W8 P=0.0053
Time point: W16 P=0.025
BMI
Time point: D1 P=0.047
<30% ≥30%

BMI, body mass index; D, day; MREG, melanoregulin; MRI-PDFF, magnetic resonance imaging-proton density fat fraction; PTEN, phosphatase and tensin homolog; W, week.

- The predictive model achieved good performance for Week 8 and even better performance for Week 16 MRI-PDFF response (accuracy = 0.88, kappa = 0.73; Table 2)

Table 2: Model Performance for Week 8 and Week 16 MRI-PDFF Response Repeated K-Fold Cross-Validation Results (K=3)

Statistics	Definition	Week 8 Model Performance	Week 16 Model Performance
KP	Accuracy improvement = (OA–EA)/(1–EA)	0.48	0.73
OA	Probability of making the right prediction	0.81	0.88
EA	Accuracy with a random classifier	0.63	0.54
SN	Probability of rejecting actual negatives	0.88	0.92
SP	Probability of detecting actual positives	0.60	0.82
PR	Probability of true positive call	0.88	0.91
NPV	Probability of true negative call	0.70	0.90

MRI-PDFF, magnetic resonance imaging-proton density fat fraction.

- Cross-validation methods used to evaluate the predictive model indicated consistent findings using different approaches (Table 3)

Table 3: Evaluation of the Predictive Model for Week 16 MRI-PDFF Response Using Different Cross-Validation Methods

Statistic	Boot	Boot632	Loocv	Repcvk3	Repcvk4	Definition
KP	0.69	0.69	0.73	0.73	0.73	Accuracy improvement = (OA–EA)/(1–EA)
OA	0.86	0.86	0.88	0.88	0.88	Probability of making the right prediction
EA	0.56	0.56	0.54	0.53	0.54	Accuracy with a random classifier
SN	0.90	0.90	0.93	0.91	0.92	Probability of rejecting actual negatives
SP	0.82	0.81	0.78	0.82	0.82	Probability of detecting actual positives
PR	0.90	0.90	0.88	0.91	0.91	Probability of true positive call
NPV	0.85	0.84	0.88	0.88	0.90	Probability of true negative call

MRI-PDFF, magnetic resonance imaging-proton density fat fraction.

Conclusions

- This work demonstrates a proof of concept for predicting which subjects will have greater (≥30%) MRI-PDFF reduction in response to AXA1125: the regression models achieved good performance at Week 16
- Protein predictors at each time point may reflect activated biological pathways relevant to an early improvement in lipid metabolism, resulting in an improvement in hepatic fat content, and are in line with the multitargeted mode of action of AXA1125
- Future efforts aim to reproduce these results and extend models to understand hepatic inflammation and fibrosis; data from the ongoing phase 2b study (NCT04880187) in NASH with fibrosis will enable exploration of predictors of biopsy improvement, including steatosis, inflammation, and fibrosis
- Potential applications include identifying responder subgroups to AXA1125 treatment on a biopsy endpoint, or potential for synergy with other compounds with complementary mechanisms of action

References

1. Vali Y, et al. *J Hepatol.* 2021;75:S564-S565.
2. Ostroff R, et al. Poster LP11 presented at: AASLD 2020.
3. Harrison SA, et al. *Am J Gastroenterol.* 2021;116:2399–2409.
4. Loomba R, et al. *Hepatology.* 2020;72:1219–1229.
5. Loomba R, et al. *Gastroenterology.* 2019;156:88–95.E5.
6. Dennis A, et al. *Front Endocrinol (Lausanne).* 2021;11. doi:10.3389/fendo.2020.575843.
7. Hamill MJ, et al. *iScience.* 2020;23:101628.

Acknowledgments

This study was supported by Axcella Therapeutics, Cambridge, MA. Medical writing support and editorial assistance were provided by Evidence Scientific Solutions, Inc. (Philadelphia, PA) and funded by Axcella Therapeutics.

Disclosures

All authors are employees of Axcella Therapeutics and may own Axcella stock/options.

Corresponding author: Karim Azer, PhD (kazer@axcellatx.com)

Presented at the American Association for the Study of Liver Diseases (AASLD) Liver Meeting, Washington, DC, USA, November 4–8, 2022