752-P

LIVRQNac (AXA1125) Enhances Insulin Sensitivity in Primary Human Hepatocytes and in Subjects With NAFLD and T2D

Seth J. Baum[1], Stephen A. Harrison[2], Nadege T. Gunn[3], Ziad H. Younes[4], Anita Kohli[5], Rashmee Patil[6], Juan P. Frías[7], Michael Hamill[8], Nadine Daou[8], Jeff Zhao[8], Manu V. Chakravarthy[8], and Margaret J. Koziel[8]

[1]Excel Medical Clinical Trials, Boca Raton, FL, United States; [2]Pinnacle Clinical Research, Live Oak, TX, United States; [3]Pinnacle Clinical Research, Austin, TX, United States; [4]Gastro One, Germantown, TN, United States; [5]Arizona Liver Health, Chandler, AZ, United States; [6]South Texas Research Institute, Edinburg, TX, United States; [7]National Research Institute, Los Angeles, CA, United States; [8]Axcella Health Inc., Cambridge, MA, United States

Poster

Axcella Virtual Café

Introduction

- Nonalcoholic fatty liver disease (NAFLD) is a multifactorial condition that is mediated by dysregulated metabolic and fibroinflammatory pathways[1]
- Insulin resistance, commonly manifested as type 2 diabetes (T2D),[2] is an important driver of NAFLD and targeting these metabolic pathways is a potential therapeutic strategy
- Endogenous metabolic modulators (EMMs), a broad set of physiologically intrinsic molecules such as amino acids, fatty acids, and other lipids, can be selectively combined to form compositions that target multiple metabolic pathways key to multifactorial liver diseases such as NAFLD
- AXA1125 is a novel, orally administered investigational EMM composition comprising 5 amino acids and a derivative: leucine, isoleucine, valine, arginine, glutamine, and N-acetyl cysteine; LIVRQNac has the same constituent components combined at different relative ratios for use *in vitro* at supraphysiologic concentrations
- A clinical study (AXA1125-002) previously assessed the safety, tolerability, and biological activity of AXA1125 in subjects with NAFLD and T2D
 - Positive directional changes in biomarkers related to liver fat, insulin sensitivity, inflammation, and fibrosis were observed;[3] these results have been supported by findings in primary human cell-based systems exposed to AXA1125[4-6]

Aims

- To investigate LIVRQNac in a nonclinical primary human hepatocyte (PHH) *in vitro* model of lipotoxicity followed by an insulin challenge, assessing effects on glucose homeostasis and insulin sensitivity
- To analyze the impact of AXA1125 on metabolic and fibroinflammatory markers in subjects with NAFLD ± T2D (AXA1125-003; NCT04073368)

Methods

Nonclinical Study

- PHHs from 3 healthy human donors were incubated in complete hepatocyte-defined medium
- Cells were switched to media containing defined custom amino acid concentrations that matched those found in healthy human plasma; media were supplemented with either (i) LIVRQNac at specified fold concentrations above the plasma level (30x for LIVRQ; N-acetyl cysteine 7.5 mM) or (ii) phosphate-buffered saline (vehicle)
- After 24 hours of pretreatment, cells were switched to media containing a lipotoxic insult referred to as free fatty acid (FFA; 0.25 mM saturated FFA [2:1 oleate: palmitate] + 1 ng/mL tumor necrosis factor-alpha), or to media lacking FFA
- Cell supernatants were collected for dipeptidyl peptidase-4 (DPP4) protein (an enzyme involved in modulation of glucagon-like peptide 1 activity and directly linked to insulin sensitivity) measurements by enzyme-linked immunosorbent assay
- For the acute insulin challenge and glucose assay, PHHs were incubated with 25 mM glucose (in an insulin-free medium), then washed and incubated in glucose-free medium supplemented with or without 10–100 nM insulin
 - Insulin-mediated Akt activation was determined by a change in the ratio of phosphorylated Akt to total Akt protein
 - After 1 hour in the insulin-free, high-glucose medium and 3 hours in the glucose-free, high-insulin (or insulin-free) medium, glucose output levels were measured

Clinical Study

- Safety, tolerability, and biological activity on liver structure and function of AXA1125 in subjects with NAFLD with and without T2D were evaluated in a 16-week, single-blind, randomized, placebo-controlled study
 - The full methodology has been previously described[7]
 - Subjects were stratified by T2D status and randomized 2:2:2:1 to receive orally administered AXA1125 24.0 g, AXA1957 13.5 g, AXA1957 20.3 g (calorie-matched and isonitrogenous to AXA1125) twice daily, or a calorie-, excipient-, and color-matched placebo 24.0 g twice daily
- Biological activity was assessed as a change from baseline in metabolically relevant biologic measures such as:
 - Glucose homeostasis: glucose and insulin in fasted states, homeostasis model assessment of insulin resistance (HOMA-IR), and glycosylated hemoglobin (HbA1c)
 - Liver fat and inflammation by magnetic resonance imaging: magnetic resonance imaging-proton density fat fraction (PDFF) and corrected T1 (cT1[8])
 - Inflammation: alanine aminotransferase (ALT)
 - Fibrosis: fibrosis-4 index (FIB-4) and N-terminal type III collagen propeptide (ProC3)
- The following key thresholds of activity, which have been correlated with histological improvement, were identified: reductions of ≥30% in MRI-PDFF, ≥17 U/L in ALT, and >80 msec in cT1[9-13]
- Here, we report results from subjects with NAFLD and T2D who received at least 1 dose of AXA1125 or placebo on Day 1

Results

Nonclinical Study

- In PHHs pretreated with supraphysiological concentrations of saturated FFAs (model system to induce lipotoxicity), LIVRQNac enhanced insulin-induced Akt phosphorylation (Figure 1A) and reduced extracellular glucose levels (Figure 1B)
 - Akt activation is demonstrated by a higher ratio of phosphorylated Akt over total Akt; phosphorylation of Akt was increased by ~80% ($p<0.001$)
 - Glucose levels in cell cuture media were reduced by ~50% ($p<0.01$)
 - These findings suggest increased insulin signaling
- LIVRQNac reduced dipeptidyl peptidase-4 (DPP4) protein levels (Figure 1C)

FIGURE 1

Effects of LIVRQNac on Akt Activation (A), Glucose Levels in the Presence of Insulin (B), and DPP4 Protein Expression (C)

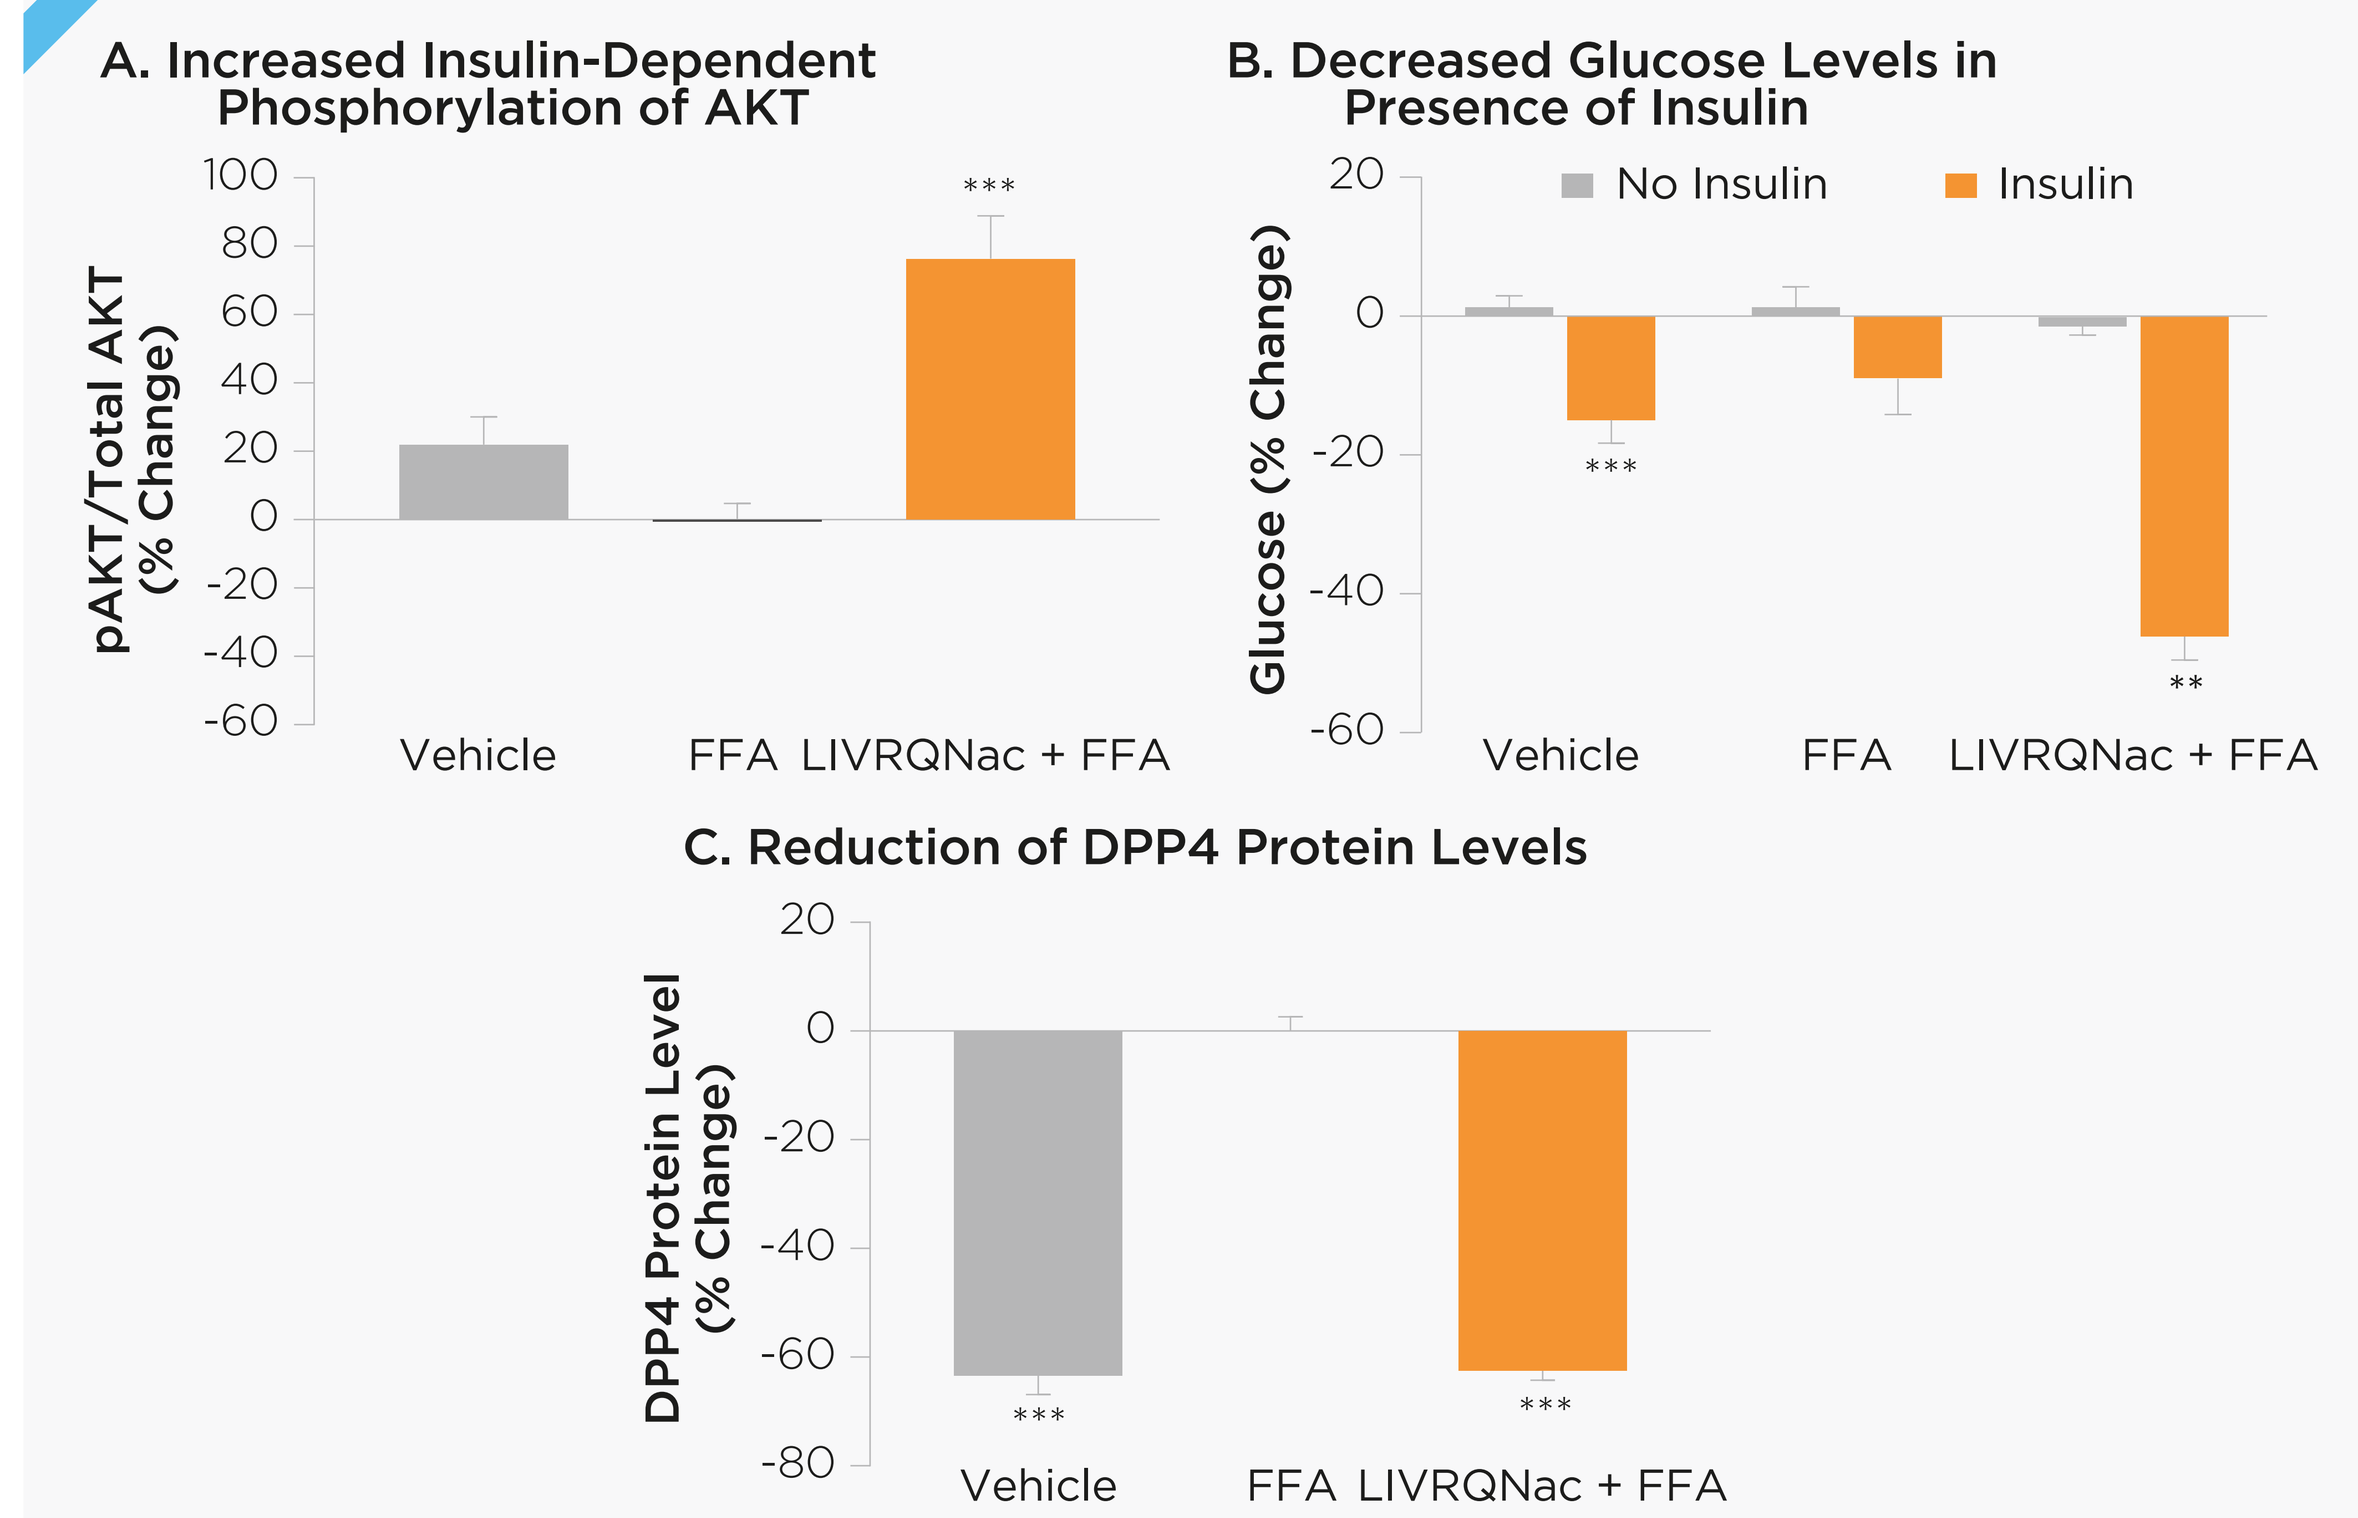

A. Increased Insulin-Dependent Phosphorylation of AKT
pAKT/Total AKT (% Change)
100
80
60
40
20
0
-20
-40
-60

Vehicle
FFA
LIVRQNac + FFA
B. Decreased Glucose Levels in Presence of Insulin
No Insulin
Insulin
Glucose (% Change)
20
0
-20
-40
-60

**
Vehicle
FFA
LIVRQNac + FFA
C. Reduction of DPP4 Protein Levels
DPP4 Protein Level (% Change)
20
0
-20
-40
-60
-80

Vehicle
FFA
LIVRQNac + FFA

p<0.01, *p<0.001 versus FFA.

DPP4, dipeptidyl peptidase 4; FFA, free fatty acids; pAkt, phosphorylated Akt.

Clinical Study

- A total of 102 subjects comprised the safety population, of which 40 (39.2%) had T2D
- Within the T2D group, 6 subjects received placebo, 12 received AXA1125
 - Baseline characteristics and demographics were generally similar among the placebo and AXA1125 groups (Table 1)

Table 1: Demographics and Baseline Characteristics in Subjects With T2D

	Placebo (n=6)	AXA1125 (n=12)
Age, years	51.7 (12.6)	51.0 (10.8)
Sex		
Female, n (%)	4 (66.7)	9 (75.0)
Weight, kg	122.4 (38.9)	101.2 (13.9)
BMI, kg/m²	41.4 (11.09)	38.6 (5.76)
Metabolism		
Fasting plasma glucose, mg/dL	130.3 (24.5)	160.9 (58.3)
Fasting plasma insulin, mIU/L	40.6 (12.0)	42.3 (44.7)
Fasting triglycerides, mg/dL	148.8 (56.1)	174.5 (104.6)
Liver fat content by MRI-PDFF, %	23.0 (5.2)	23.6 (4.5)
HOMA-IR	12.9 (3.6)	19.2 (25.8)
HbA1c, %	6.9 (0.5)	7.8 (0.9)
Inflammation		
ALT, U/L	51.3 (42.3)	60.8 (26.2)
cT1, msec	1040.0 (204.7)	999.3 (105.1)
Fibrosis		
FibroScan score, kPa	14.2 (5.7)	9.9 (1.3)
ProC3, ng/mL	13.8 (4.0)	18.3 (10.6)
ELF score	9.2 (1.3)	9.4 (0.7)
FIB-4	1.1 (0.3)	1.2 (0.8)

All values are mean (SD) unless otherwise noted.

ALT, alanine aminotransferase; BMI, body mass index; cT1, corrected T1; ELF, Enhanced Liver Fibrosis; FIB-4, fibrosis 4; HbA1c, glycated hemoglobin; HOMA-IR, homeostasis model assessment of insulin resistance; MRI-PDFF, magnetic resonance imaging-proton density fat fraction; ProC3, N-terminal type III collagen propeptide; SD, standard deviation; T2D, type 2 diabetes.

Biological Activity

- At Week 16, AXA1125 demonstrated consistently greater activity versus placebo across biomarkers of metabolism and fibroinflammation in subjects with T2D (Figures 2–4)
- For fasting glucose (Figure 2A) and fasting insulin (Figure 2B), larger absolute reductions from baseline were observed with AXA1125 compared with placebo
- Administration of AXA1125 also led to larger mean reductions from baseline in HOMA-IR (Figure 2C) and HbA1c (Figure 2D) compared with placebo

FIGURE 2

Reduction From Baseline in Biomarkers of Glucose Homeostasis at Week 16 in (A) Fasting Glucose, (B) Fasting Insulin, (C) HOMA-IR, and (D) HbA1c

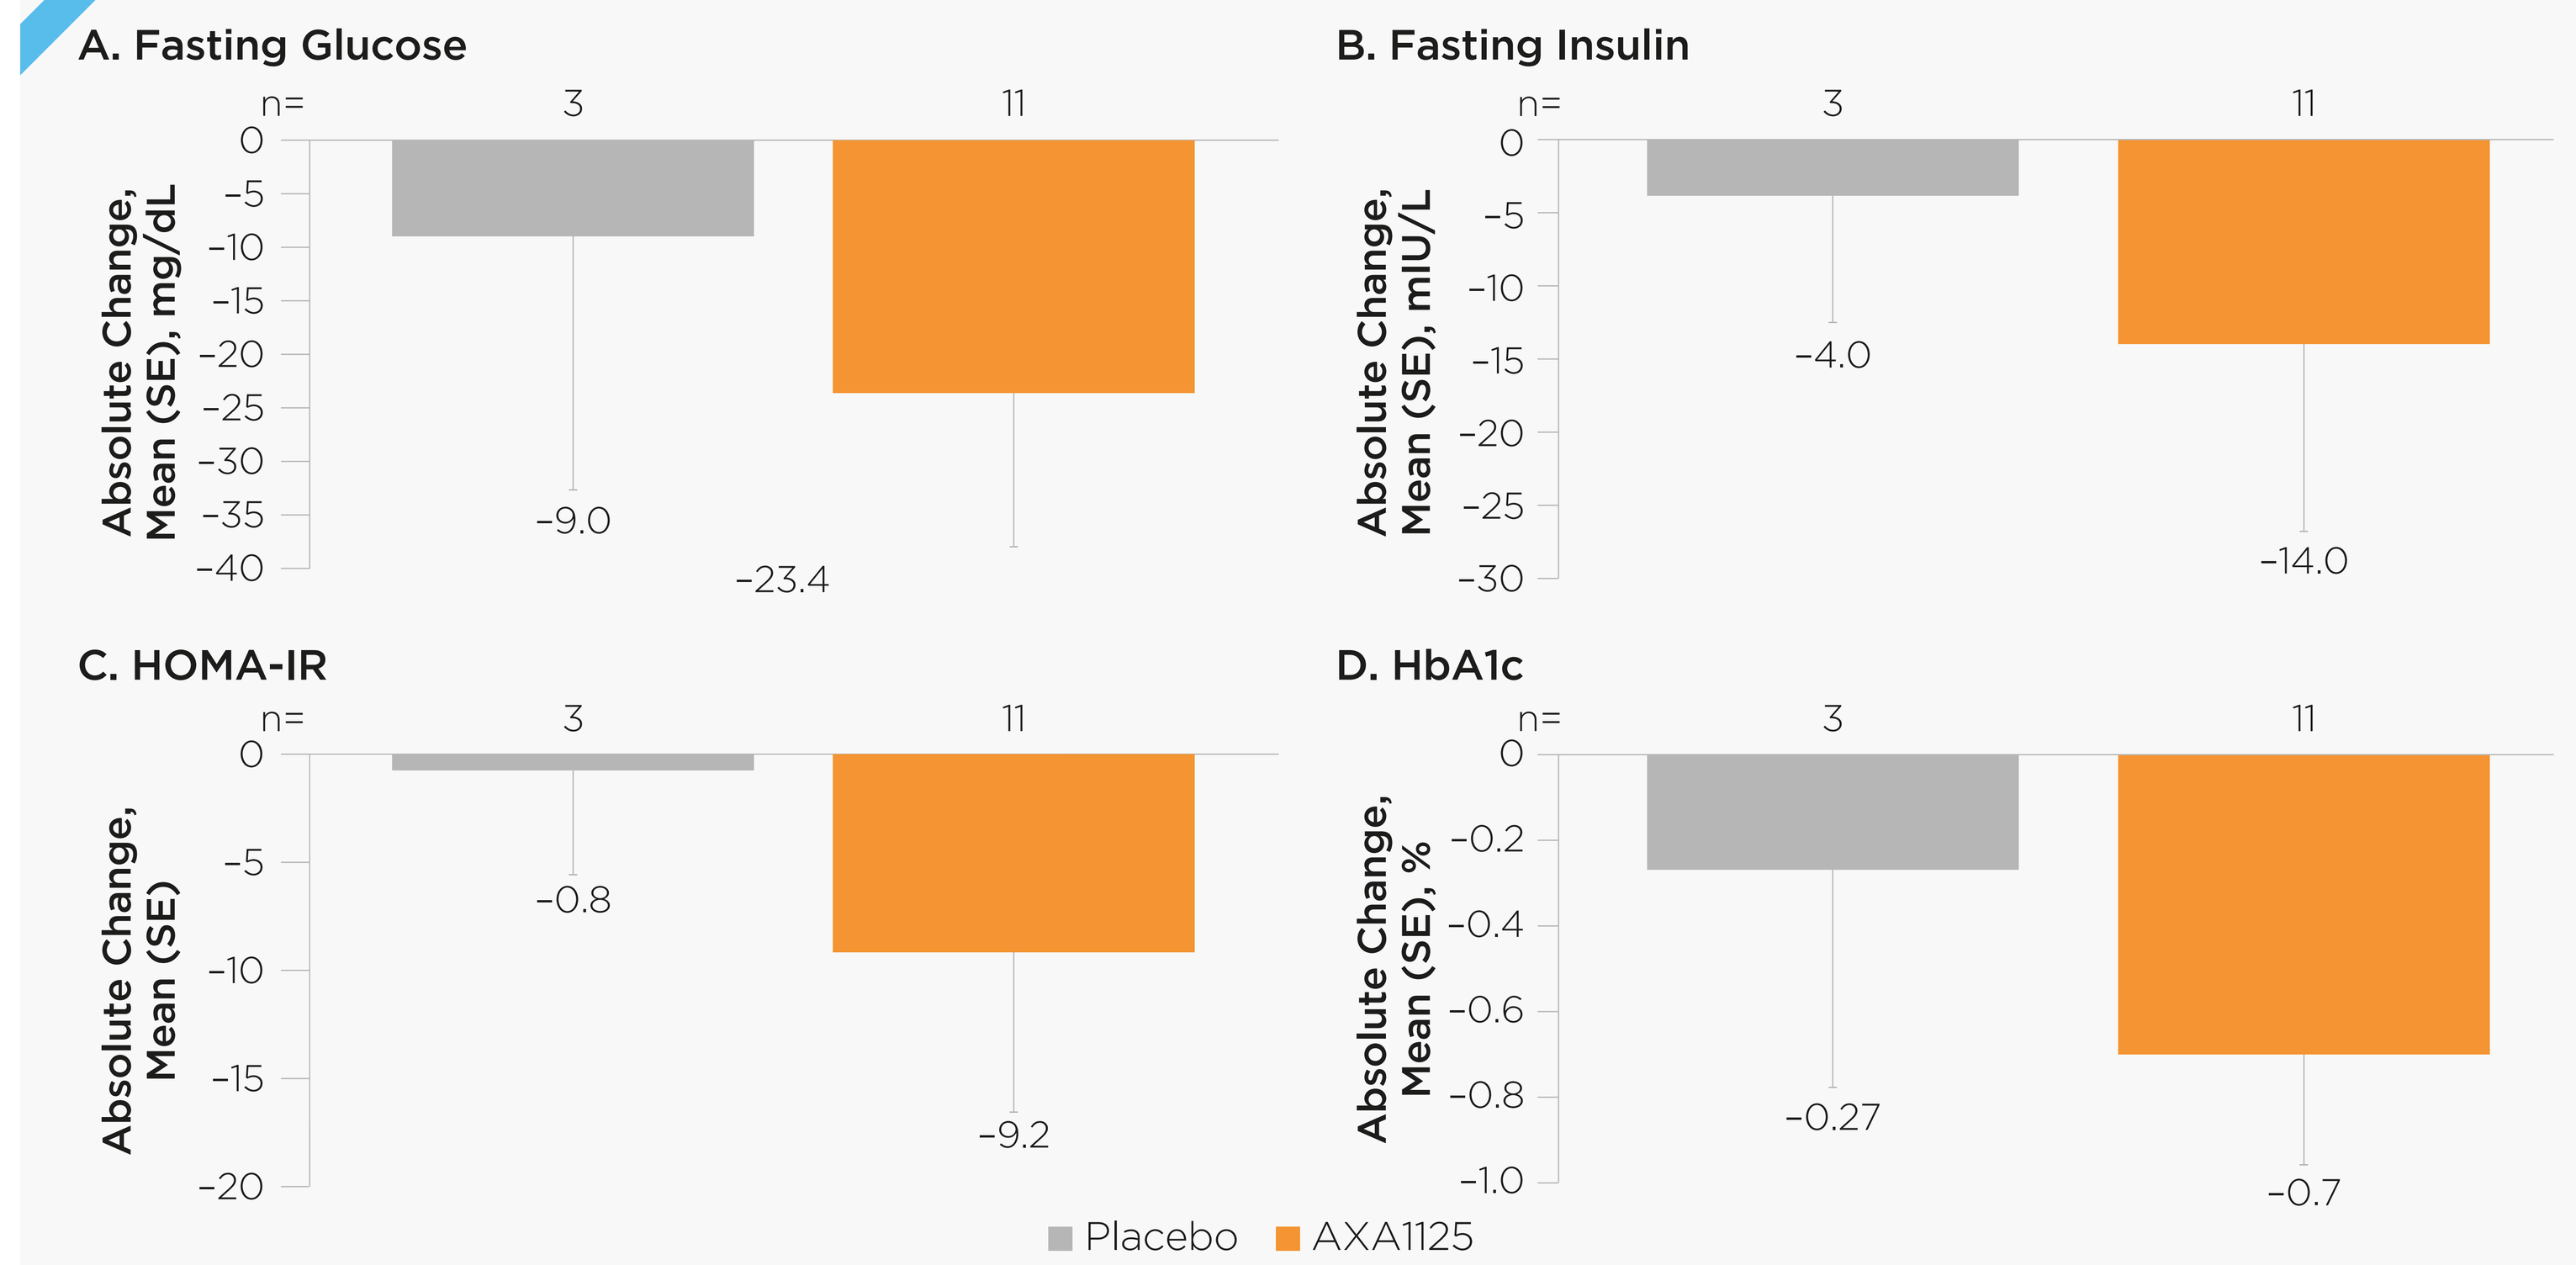

A. Fasting Glucose
n= 3 11
Absolute Change, Mean (SE), mg/dL
-9.0
-23.4
B. Fasting Insulin
n= 3 11
Absolute Change, Mean (SE), mIU/L
-4.0
-14.0
C. HOMA-IR
n= 3 11
Absolute Change, Mean (SE)
-0.8
-9.2
D. HbA1c
n= 3 11
Absolute Change, Mean (SE), %
-0.27
-0.7
Placebo
AXA1125

HbA1c, glycated hemoglobin; HOMA-IR, homeostatic model assessment of insulin resistance; SE, standard error; T2D, type 2 diabetes.

- At Week 16, there was a larger mean relative reduction from baseline in MRI-PDFF with AXA1125 compared with placebo in subjects with T2D (Figure 3A)
- Compared with the placebo group, the AXA1125 group had more pronounced changes from baseline in ALT (Figure 3B), cT1 (Figure 3C), and ProC3 (Figure 3D)
- While a meaningful relative reduction in FIB-4 was seen from baseline to Week 16 in the AXA1125 group, a larger reduction was seen in the placebo group (Figure 3E)

FIGURE 3

Reduction From Baseline at Week 16 in Biomarkers of Metabolism A) MRI-PDFF, Inflammation (B) ALT and (C) cT1, and Fibrosis (D) ProC3 and (E) FIB-4

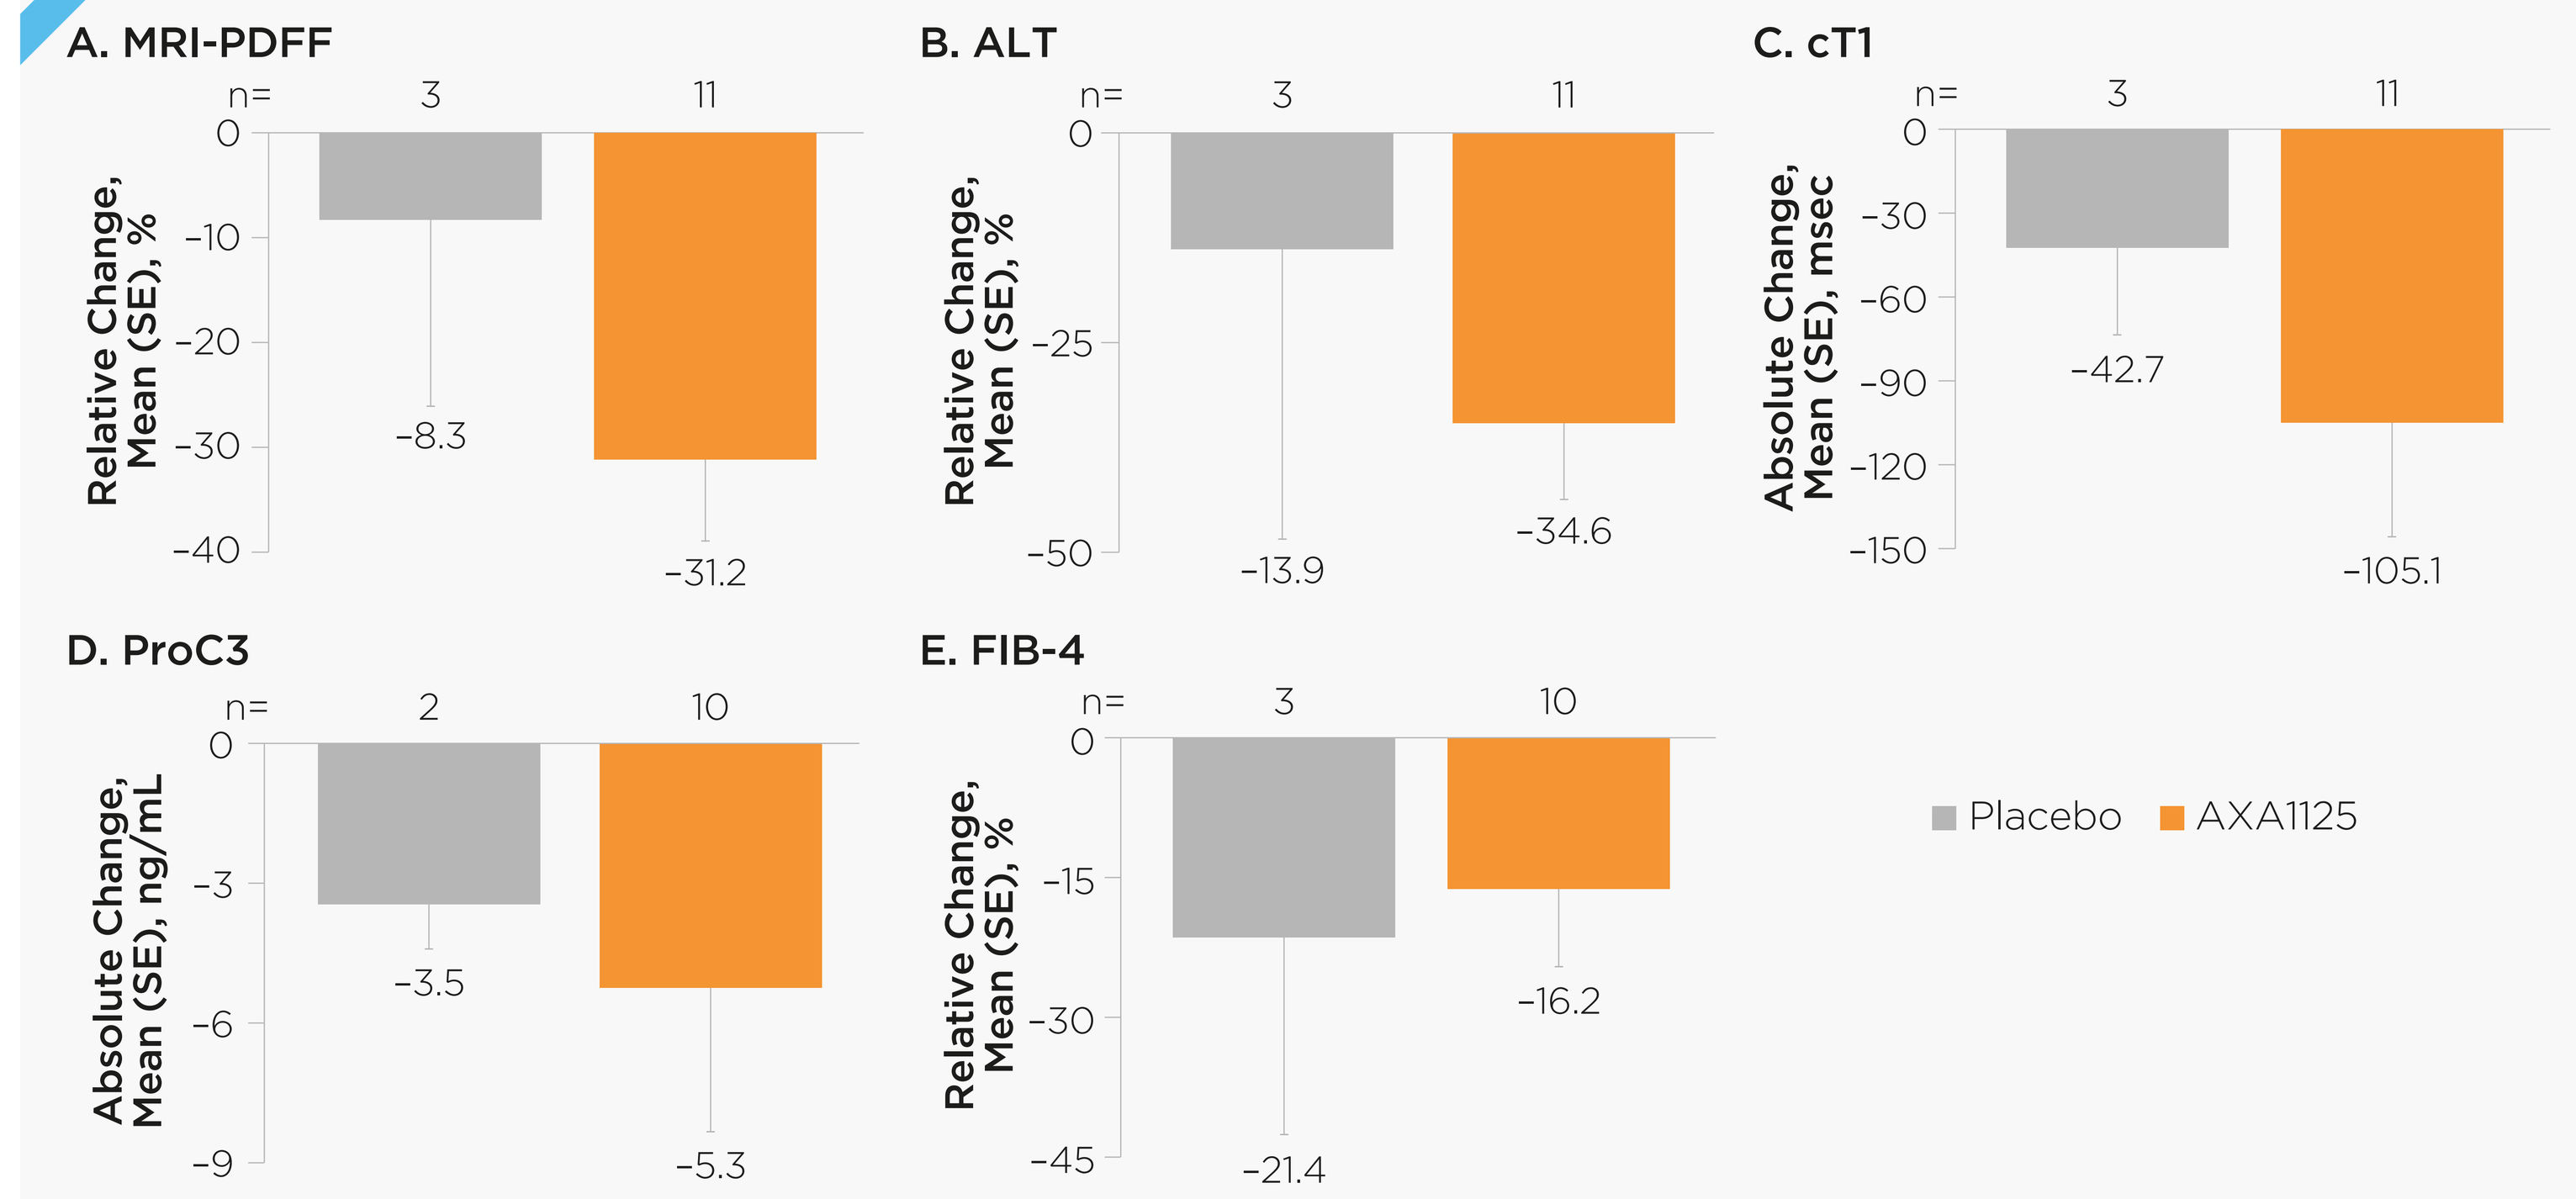

A. MRI-PDFF
n= 3 11
Relative Change, Mean (SE), %
-8.3
-31.2
B. ALT
n= 3 11
Relative Change, Mean (SE), %
-13.9
-34.6
C. cT1
n= 3 11
Absolute Change, Mean (SE), msec
-42.7
-105.1
D. ProC3
n= 2 10
Absolute Change, Mean (SE), ng/mL
-3.5
-5.3
E. FIB-4
n= 3 10
Relative Change, Mean (SE), %
-21.4
-16.2
Placebo
AXA1125

ALT, alanine aminotransferase; cT1, corrected T1; FIB-4, fibrosis-4; MRI-PDFF, magnetic resonance imaging proton density fat fraction; ProC3, N-terminal type III collagen propeptide; SE, standard error; T2D, type 2 diabetes.

- There is increasing evidence linking a >80 msec absolute cT1 reduction, 30% relative reduction in MRI-PDFF and 17 U/L absolute reduction in ALT with improved histologic outcomes[9-13]
- At Week 16, 46%–64% of subjects with T2D who received AXA1125 achieved at least an 80 msec absolute reduction in cT1, 30% relative reduction in MRI-PDFF, or 17 U/L absolute reduction in ALT (Figures 4A, 4B, and 4C)
- The percentage of subjects achieving these thresholds with AXA1125 was up to ~1.7-fold higher in the T2D population compared with the overall population

FIGURE 4

Subjects With T2D Achieving Meaningful Reduction Thresholds Associated With Improved Histologic Outcomes at Week 16

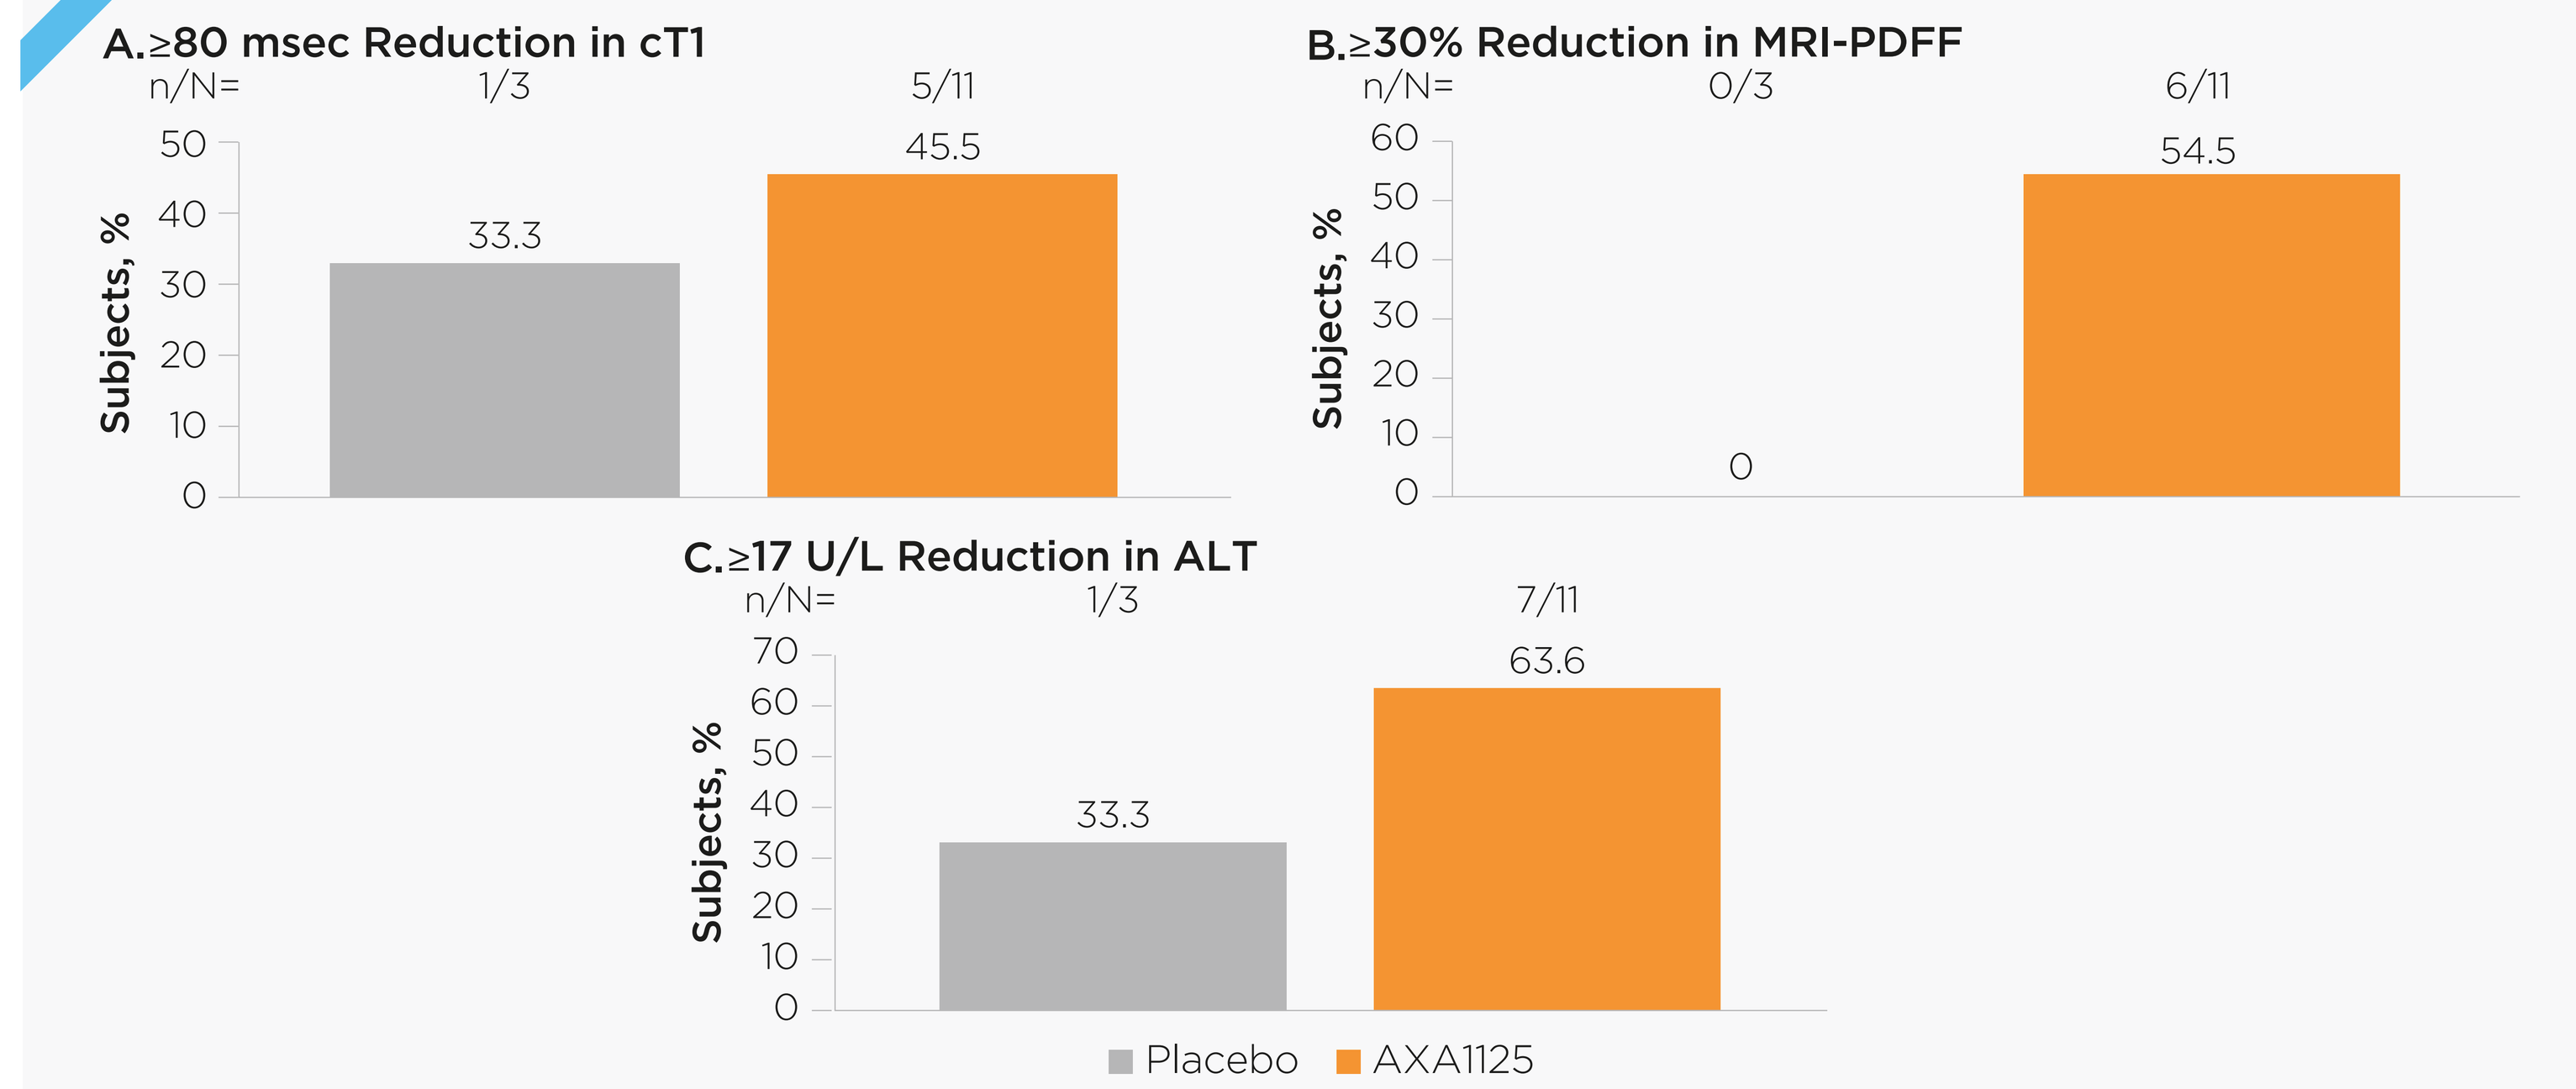

A. ≥80 msec Reduction in cT1
n/N= 1/3 5/11
Subjects, %
33.3
45.5
B. ≥30% Reduction in MRI-PDFF
n/N= 0/3 6/11
Subjects, %
0
54.5
C. ≥17 U/L Reduction in ALT
n/N= 1/3 7/11
Subjects, %
33.3
63.6
Placebo
AXA1125

ALT, alanine aminotransferase; cT1, corrected T1; MRI-PDFF, magnetic resonance imaging-proton density fat fraction; T2D, type 2 diabetes.

Safety

- In the overall population, AXA1125 was generally well tolerated, with no serious AEs related to study product
- Four subjects had AEs that led to study discontinuation (1 with placebo, 1 with AXA1125, and 2 with AXA1957 high dose)
- Product-emergent AEs for those administered AXA1125 were mostly mild to moderate, with only 1 severe AE reported (in the placebo group) and no clinically meaningful differences in serum lipids or body weight were observed

Conclusions

- In the PHH model, LIVRQNac improved glucose homeostasis and enhanced insulin sensitivity
- In the clinical study, concordant changes in multiple measures of biologic activity were consistent with improvements in glucose homeostasis, reductions in liver fat, and decreases in fibroinflammation
 - Glucose, insulin, HOMA-IR, HbA1c, and markers of liver fat and fibroinflammation improved with AXA1125 compared with placebo
 - Results with AXA1125 administration from this study are consistent with those from a previous clinical study in T2D subjects with NAFLD,[3] and phenotypic and mechanistic data[4-6]
- A phase 2b clinical trial (NCT04880187; EMMPACT) of AXA1125 is currently actively enrolling F2/F3 patients with nonalcoholic steatohepatitis (NASH) with and without T2D

References

1. Friedman SL, et al. *Nat Med.* 2018;24:908–922.
2. Mu W, et al. *Front Pharmacol.* 2019;9:1566.
3. Marukian S, et al. *Hepatology.* 2018;68(Suppl 1):67A. Abstract 106.
4. Chakravarthy MV, et al. *Hepatology.* 2019;70(Suppl 1):1264A. Abstract 2134.
5. Lee CW, et al. Poster P02-05 presented at: EASL NAFLD Summit 2017.
6. Marukian S, et al. Oral presentation at: Keystone Symposia 2019.
7. Harrison SA, et al. Poster presented at: The Digital International Liver Congress 2020.
8. Pavlides M, et al. *Liver Int.* 2017;37:1065-1073.
9. Loomba R, et al. *Hepatology.* 2020;72:1219–1229.
10. Dennis A, et al. *Front Endocrinol.* 2021;11:575843.
11. Loomba R, et al. *Gastroenterology.* 2019;156:88–95.e5.
12. Harrison SA, et al. *Hepatology.* 2020;71:1198–1212.
13. McDonald N, et al. *Sci Rep.* 2018;8:9189.

Acknowledgments

Medical writing support and editorial assistance were provided by Evidence Scientific Solutions, Inc (Philadelphia, PA). This support was funded by Axcella Health Inc.

Disclosures

SJB: Consultant: Akcea, Amgen, Novo Nordisk, Regeneron, Sanofi; Sponsored lectures: Amgen, Boehringer Ingelheim, Eli Lilly and Company, Novo Nordisk; Scientific advisory board: Akcea, Amgen, AstraZeneca, Esperion, Novartis, Regeneron, Sanofi. **SAH:** Stock ownership or equity: Akero, Cirius, Galectin, Genfit, HistoIndex, Madrigal, Metacrine, NGM Biopharmaceuticals, NorthSea; Consulting/Advisory: Akero, Altimmune, Axcella Health, Blade Therapeutics, Cirius, CiVi Biopharma, CLDF, CymaBay, Echosens, Foresite Labs, Galectin, Galmed, Gelesis, Genfit, Gilead, HighTide, HistoIndex, Hepion, Indalo, Innovate, Intercept, Madrigal, Medpace, Metacrine, NGM Biopharmaceuticals, NorthSea, Novartis, Novo Nordisk, Perspectum, Poxel, ProMetic Life Sciences, Ridgeline Therapeutics, Sagimet Biosciences, Terns, Viking; Grants for clinical research: Axcella Health, Bristol Myers Squibb, Cirius, CiVi Biopharma, Conatus, CymaBay, Enyo Pharma, Galectin, Galmed, Genentech, Genfit, Gilead, Hepion, HighTide, Immuron, Intercept, Madrigal, Metacrine, NGM Biopharmaceuticals, NorthSea, Novartis, Novo Nordisk, Pfizer, Sagimet Biosciences, Second Genome, Tobira/Allergan, Viking. **NTG:** Research grants: Axcella Health, BMC Pharma, CymaBay, Genentech, Genfit, Gilead, HighTide, Madrigal, NorthSea, NGM Biopharmaceuticals, Novo Nordisk; Consultant/Speaker: AbbVie, Dova, Gilead, Intercept, Salix. **ZHY:** Grants: AXA, Bristol Myers Squibb, CymaBay, Gilead, Intercept, Madrigal, NGM Biopharmaceuticals, Novo Nordisk, Zydus; Consultant: Gilead; Sponsored lectures: Intercept. **AK:** Grants: Gilead; Consultant: Gilead, Intercept, Novartis. **RP:** Nothing to disclose. **JPF:** Research support: AbbVie, Akero, AstraZeneca, BMS, Boehringer Ingelheim, Eli Lilly, Intercept, Janssen, Madrigal, Metacrine, Merck, NorthSea, Novartis, Novo Nordisk, Oramed, Pfizer, Poxil, Sanofi, Theracos; Advisory boards and consulting: Akero, Altimmune, Axcella Health, Boehringer Ingelheim, Coherus Therapeutics, Echosens, Eli Lilly, Gilead, Intercept, Merck, Novo Nordisk, Sanofi, 89bio; Speaker Bureau: Eli Lilly, Merck, Sanofi. **MH, JZ, MJK:** Employees of Axcella Health and may own stock options in the company. **ND, MVC:** Employees of Axcella Health during the time the studies were conducted.

Corresponding author: Margaret J. Koziel, MD (mkoziel@axcellahealth.com)